A Novel Two-step Ionic and Acid Dissociation of Drug, Anti-drug Antibody Complexes for Use in Immuno-assays.

Stacy Sandmeier, Melissa Taylor, LaHoma Easterwood, John Pirro, Jackie Dillon

KCAS bio
SCIENCE ACCELERATED

INTRODUCTION

Drug Tolerance is a critical hurdle for most anti-drug antibody (ADA) assay development. The most commonly used method to remove bound drug from an ADA is through acid dissociation. Unfortunately, acid dissociation has limitations since strong acids can denature ADAs. Alternatively, High ionic strength dissociation assays (HISDA) as introduced by Jordan et al 1 does increase drug tolerance for most assays over acid dissociation with little to no signal loss. However, due to the gentle non-denaturing effect of MgCl2 tightly bound hydrophobic drug-anti-drug antibodies do not dissociate. Hydrophobic interactions (protein folding of the drug or ADA), may prevent the acid or MgCl2 access to the bonds between the drug and the antibody. We report here the use of MgCl2 prior to the addition of an acid to create an increased ionic strength solution and salt precipitation. The ionic solution temporarily changes the folding structures of the drug and or bound antibodies cleaving the drug-anti-drug antibody bond, thereby releasing the ADA from drug complexes. This improves drug tolerance and makes the free ADA more available for antibody detection in a variety of assays.

METHOD

Drug Tolerance assay: Here we describe a two-step acid and ionic dissociation method using salt precipitation to treat samples potentially containing ADA complexes. Samples containing drug and anti-drug antibody complexes are combined in equal volume with MgCl2 (concentration is driven by ADA stability) for a 30-40 minute incubation at room temperature while shaking. MgCl2 solution, by itself, is at a neutral pH, is non-denaturing, and should cause minimal changes to serum proteins' secondary and tertiary structures1. An acid with a pH of approximately 2.5, such as 400 mM Glycine or 300 mM Acetic Acid (or another acceptable acid), is then added to the samples containing MgCl2 at appropriate volumes to bring the final sample pH into the acidic range. The samples are then incubated an additional 30-40 minutes (acid time is dependent on antibody stability and the range may need to be adjusted accordingly). Bringing the MgCl2 into acidic range increases the ionic strength of the solution. The high ionic strength reduces the solubility of some serum proteins, thereby causing a salt precipitate. This salt precipitate has been shown to be mostly albumins and has little effect on the anti-drug antibody itself. However, the increase in ionic strength causes reversible folding alterations. Dissociation can now take place between the drug and the ADAs more completely, freeing more ADAs to be determined in subsequent testing. When the pH returns to the neutral range and the MgCl2 is diluted, the salt precipitated matrix components go back into solution. Neutralization occurs in conjunction with a free drug capture step such as an ACE method or bead binding method to remove the newly unbound drug from the system.

Salt precipitation: the method used for the drug tolerance assays creates a salt precipitation not long after the addition of Acid at pH 2.5 to the MgCl2 sample dilution. In order to test what was precipitating and if the precipitate went fully back into solution, the following method was used. In a non-binding plate samples containing either BSA, serum, plasma, sucrose, ovalbumin, 1x PBS, glucose, fibrogen, fibrogen extracted matrix, NaCl, histidine, or IgG (human IgG1-4) were added at equal volumes to 2 M MgCl2. To this solution 400 mM Glycine was added at 1:2.5 dilution for a total of 1:5 dilution. The plate(s) were read at 490, 540, 656 and 280 nm as a kinetic read every 5 minutes for 30 minutes. After the precipitate was formed 250 mM Tris +3% BSA, pH 8.0 was added to return the solution to a neutral pH range. Kinetic reads at 490, 540, 656 and 280 nm were performed every 5 minutes for 30 minutes.

RESULTS

Salt precipitation: Precipitation occurred at wavelengths of 490, 540 and 656 nM for BSA, Ovalbumin, serum and plasma. IgG concentration curves ranged from 10 mg/mL to 0.0313 mg/mL and was tested in the presence of 1X PBS no dissociation, 400 mM Glycine dissociation only, 2 M MgCl2 dissociation only and 2 M MgCl2 followed by 400 mM Glycine dissociation as described above. No change occurred in the wavelength between the different assay conditions. IgG samples showed no increase in signal for any of the wavelengths tested and no visual precipitation occurred.

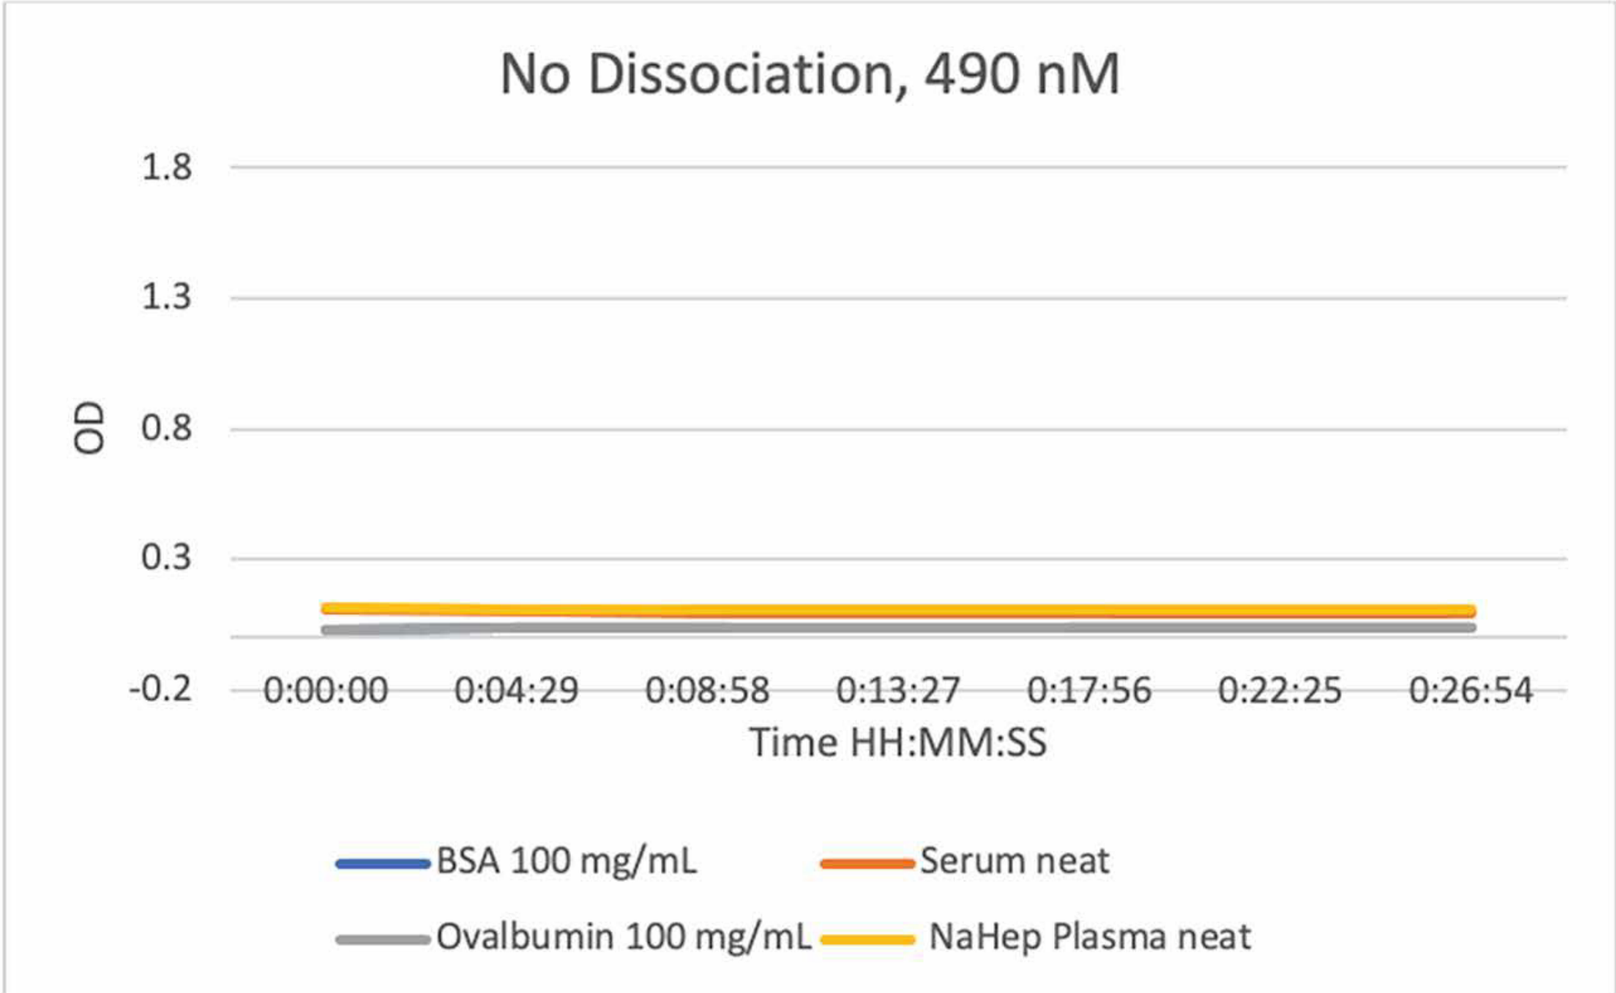
No Dissociation, 490 nM
OD
1.8
1.3
0.8
0.3
-0.2
0:00:00
0:04:29
0:08:58
0:13:27
0:17:56
0:22:25
0:26:54
Time HH:MM:SS
BSA 100 mg/mL
Serum neat
Ovalbumin 100 mg/mL
NaHep Plasma neat

Figure 1: 1XPBS used to dilute the samples, no dissociation applied to samples tested. No precipitation noted in the wells.

Base line values for the samples used were below 0.3 OD units at 490 nm. 540 and 656 nm shared lower but similar signal values to 490 nm.

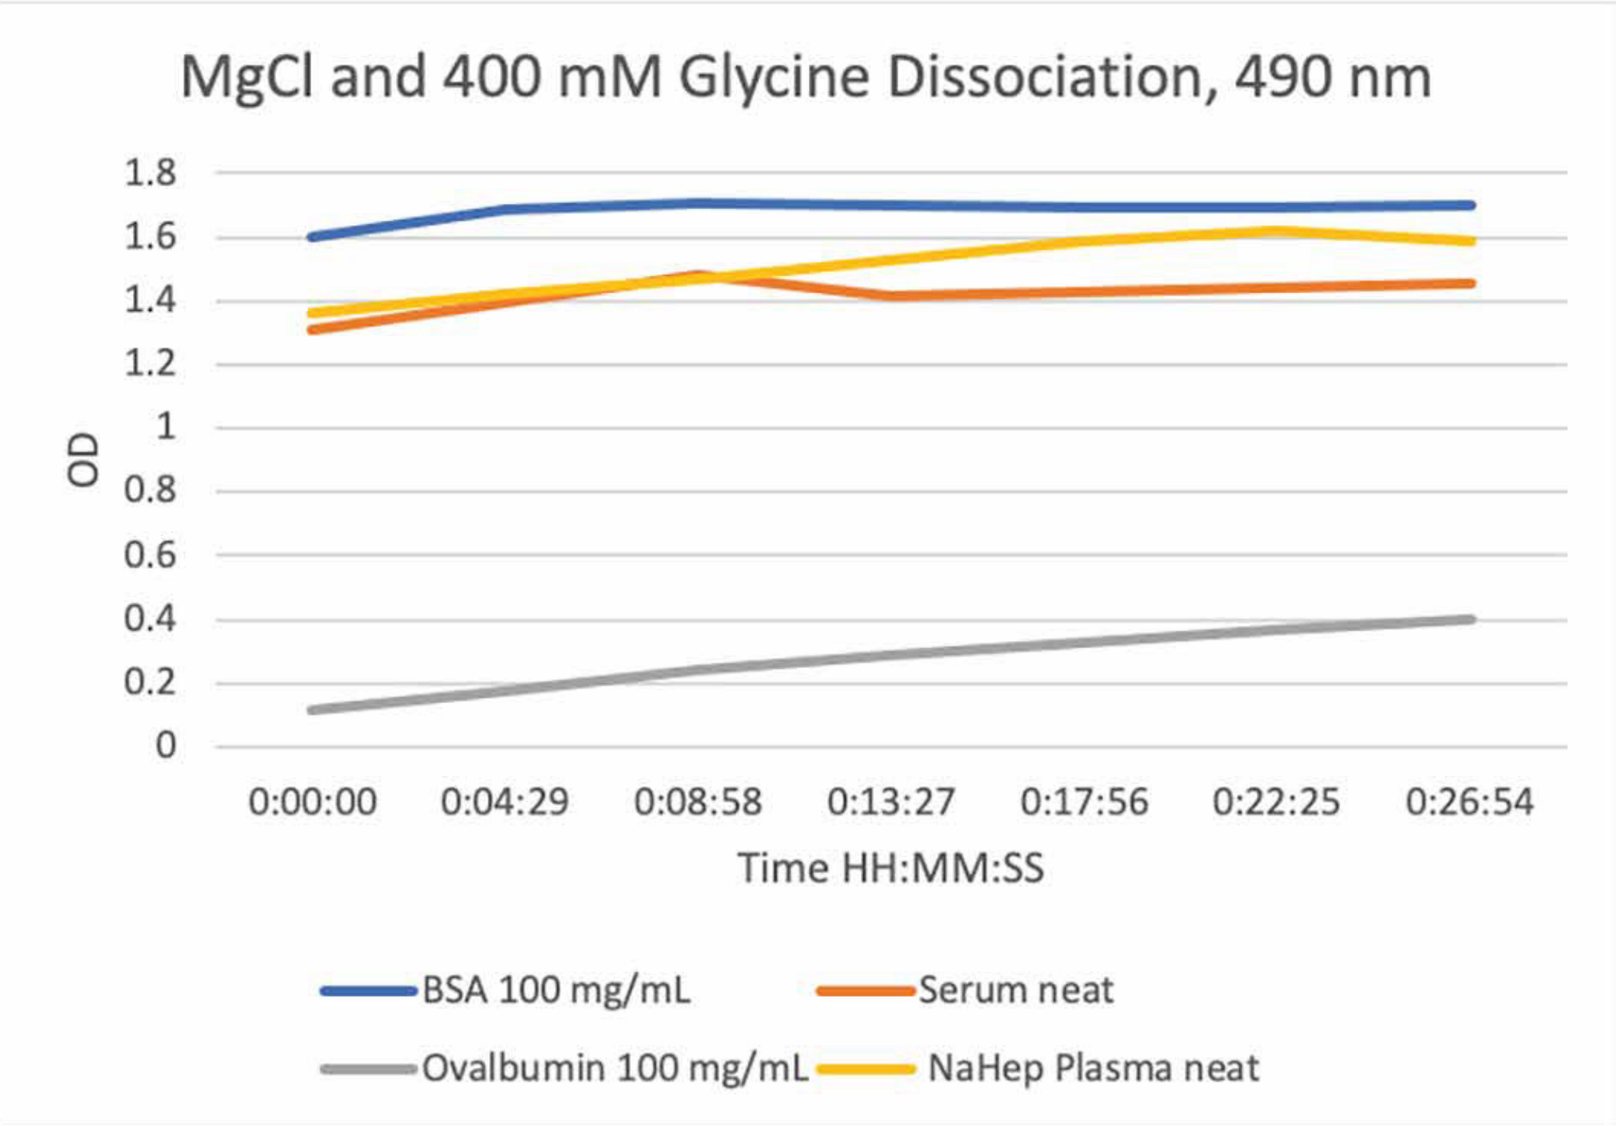
MgCl and 400 mM Glycine Dissociation, 490 nm
OD
1.8
1.6
1.4
1.2
1
0.8
0.6
0.4
0.2
0
0:00:00
0:04:29
0:08:58
0:13:27
0:17:56
0:22:25
0:26:54
Time HH:MM:SS
BSA 100 mg/mL
Serum neat
Ovalbumin 100 mg/mL
NaHep Plasma neat

Figure 2: Sample dissociation using 2M MgCL2 treatment, followed by 400mM Glycine. Full precipitation occurred prior to the end of the sample dissociation incubation.

Precipitation began occurring immediately upon acid addition, prior to the start of the kinetic read at 490, 540 and 656 nm (490 nm shown). Once Neutralization buffer was added signal began to immediately drop back to base line values.

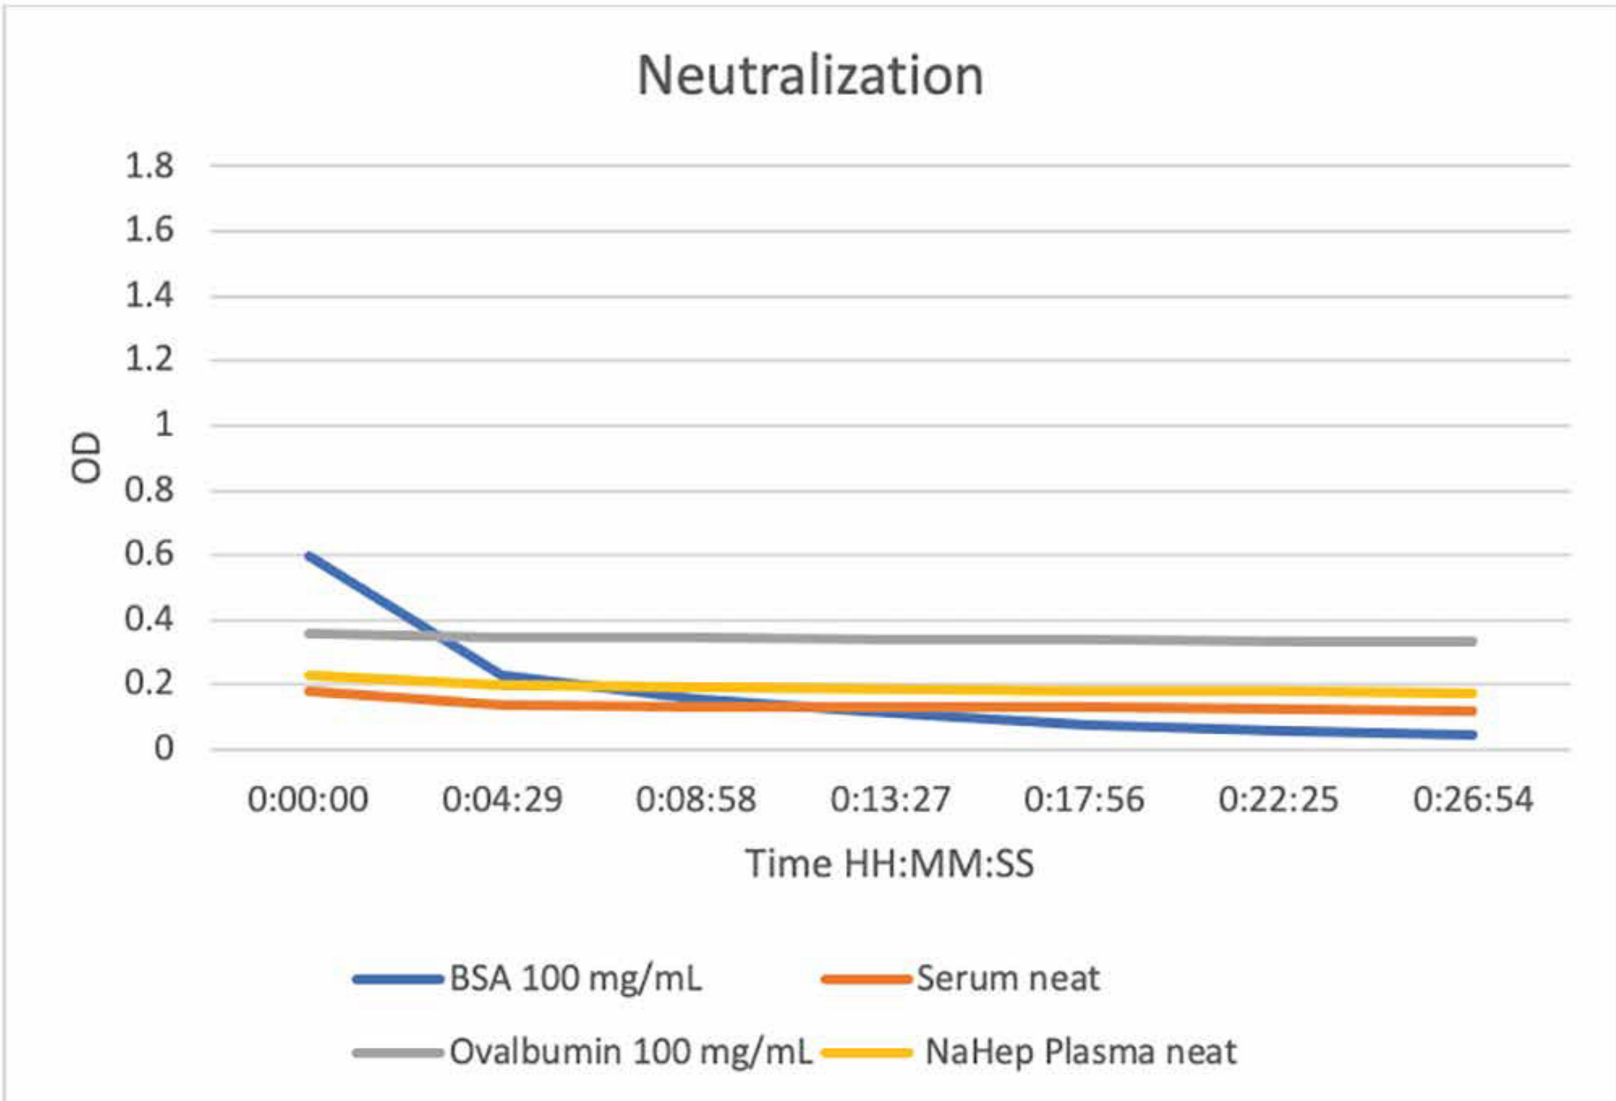
Neutralization
OD
1.8
1.6
1.4
1.2
1
0.8
0.6
0.4
0.2
0
0:00:00
0:04:29
0:08:58
0:13:27
0:17:56
0:22:25
0:26:54
Time HH:MM:SS
BSA 100 mg/mL
Serum neat
Ovalbumin 100 mg/mL
NaHep Plasma neat

Figure 3: Precipitation pellets went back into solution within 5 minutes of the addition of neutralization buffer. OD values of samples tested fell at or below values seen for the no dissociation samples once the precipitation pellet was re-suspended.

Drug Tolerance: Multiple experiments with a non-drug tolerant monoclonal anti-drug antibody were performed using 3 M MgCl2 in comparison to the acetic acid dissociation in the original method. The assay was performed using a 1:5 dilution of sample in either 300 mM Acetic Acid or 3 M MgCl2 for a 30 minute incubation prior to neutralization (250 mM Tris, pH 8.0 for the acetic acid and 1X PBS+Blocking buffer for the MgCl2). The MgCl2 dissociation did have a higher RLU signal overall for the antibody, as expected based on the HISDA paper by Jordan et. al 1; however, we did not have a significant increase in drug tolerance using MgCl2 alone. It is of note that the HISDA method uses 4 M MgCl2. In our laboratory we have found that high concentrations of MgCl2 caused significant loss in antibody signal overtime in this assay. Thus selection of the appropriate MgCl2 concentration is critical for assay development.

MRD at 1:5: Signal to noise

Sample	300 mM Acetic Acid	3 M $MgCl_2$
100 ng/mL ADA + 200 µg/mL Drug	0.7	0.8
100 ng/mL ADA + 50.0 µg/mL Drug	0.8	1.1
100 ng/mL ADA + 10.0 µg/mL Drug	1.0	1.4
100 ng/mL ADA + 0.0 µg/mL Drug	1.9	3.0

Table 1: Acid Dissociation vs MgCL2 dissociation in Monoclonal antibody assay. MRD at 1:5: Data represented as signal to noise

Blue shading indicates the sample is above the cutpoint's signal-to-noise ratio for the assay.

The low drug tolerance led to the development of a stepwise procedure treating first with MgCl2 then with Acid (developed in multiple assays with differing acids, drugs, and ADAs). Drug tolerance was improved from none to minimal drug tolerance up to concentrations of 1000 µg/mL for the drug evaluated depending on antibody concentration and method of removing the unbound drug.

No MRD: Final assay as listed in Method section: Signal to noise

Sample	3 M $MgCl_2$ + 300 mM Acetic Acid
100 ng/mL ADA + 1000 µg/mL Drug	2.0
100 ng/mL ADA + 400 µg/mL Drug	2.3
100 ng/mL ADA + 200 µg/mL Drug	2.2
100 ng/mL ADA + 100 µg/mL Drug	2.6
100 ng/mL ADA + 50.0 µg/mL Drug	2.5
100 ng/mL ADA + 20.0 µg/mL Drug	2.7
100 ng/mL ADA + 10.0 µg/mL Drug	2.8
100 ng/mL ADA + 0.0 µg/mL Drug	31.6
NC	1.0

Table 2: No MRD prior to dissociation: Final assay: Data represented as signal to noise

The final optimized assay using the method as described in the method section raised the assays drug tolerance at 100ng/mL ADA to 1000µg/mL.

Blue shading indicates the sample is above the cutpoint's signal-to-noise ratio for the assay.

Monoclonal Drug Tolerance Summary			
Evaluation	**Specification**	**No Dissociation**	**$MgCl_2$ and Acid Dissociation**
Drug tolerance Comparison of No Dissociation and stepwise $MgCl_2$ and Acid dissociation	Highest tested drug:PC ratio which failed to inhibit 100.0 ng/mL anti-drug Ab below cut point.	<10.0 µg/mL	>1000 µg/mL

Table 3: Monoclonal Drug Tolerance Summary

In parallel to the monoclonal antibody assay a polyclonal assay was developed, using 1:1 dilution of sample in 2 M MgCl2 prior to a 1:2.5 dilution in Glycine for a final dilution of 1:5. Samples in the following tables that are listed as having either "No MgCl2" or "No Acid Dissociation" were treated with 1X PBS only for those steps.

Samples	Acid Dissociation only	2 M $MgCl_2$ With Acid dissociation	No Dissociation	2M $MgCl_2$ Dissociation only
200 ng/mL ADA and 100 µg/mL Drug	1.0	1.2	0.9	1.0
200 ng/mL ADA and 50.0 µg/mL Drug	1.2	1.6	1.0	1.0
200 ng/mL ADA and 10.0 µg/mL Drug	1.9	3.2	1.2	1.2
200 ng/mL ADA and 0.0 µg/mL Drug	8.1	7.2	4.2	4.0
100 ng/mL ADA and 100 µg/mL Drug	1.0	1.1	0.9	0.9
100 ng/mL ADA and 50.0 µg/mL Drug	1.1	1.3	1.0	0.9
100 ng/mL ADA and 10.0 µg/mL Drug	1.5	2.2	1.0	1.0
100 ng/mL ADA and 0.0 µg/mL Drug	4.3	4.2	2.4	2.3

Table 4: Polyclonal Drug Tolerance various dissociation solutions.

50 ng/mL ADA and 100 µg/mL Drug	0.9	1.0	1.0	0.9
50 ng/mL ADA and 50.0 µg/mL Drug	1.0	1.1	1.0	1.0
50 ng/mL ADA and 10.0 µg/mL Drug	1.2	1.3	1.0	1.0
50 ng/mL ADA and 0.0 µg/mL Drug	2.7	2.3	1.7	1.6
0 ng/mL ADA and 100 0µg/mL Drug	1.0	1.0	0.9	1.0
0 ng/mL ADA and 50.0 µg/mL Drug	1.0	1.1	0.9	1.0
0 ng/mL ADA and 10.0 µg/mL Drug	0.9	1.0	0.9	1.0
0 ng/mL ADA and 0.0 µg/mL Drug	1.0	1.0	1.0	1.0

Table 4 (continued): Polyclonal Drug Tolerance various dissociation solutions.

Blue shading indicates the sample is above the cutpoint's signal-to-noise ratio for the assay.

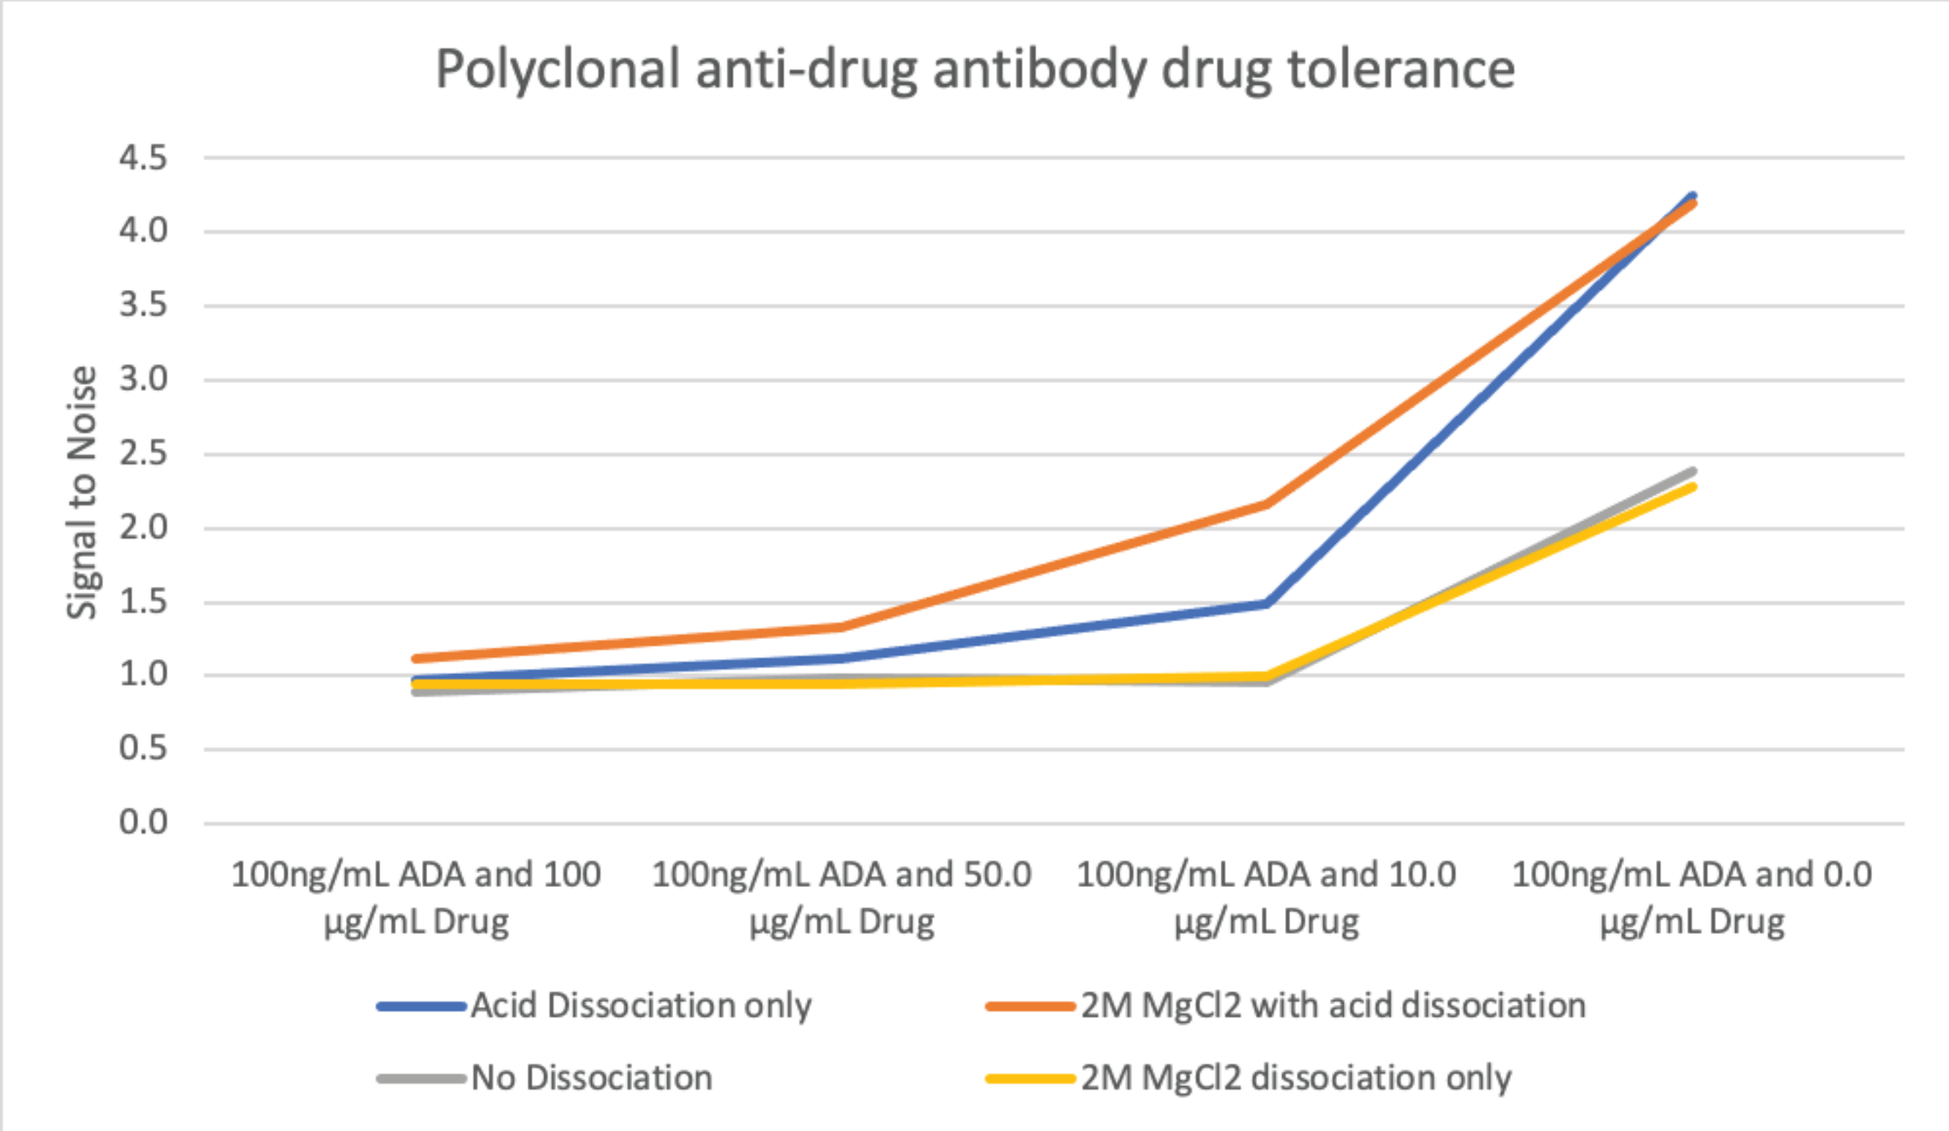
Polyclonal anti-drug antibody drug tolerance
Signal to Noise
4.5
4.0
3.5
3.0
2.5
2.0
1.5
1.0
0.5
0.0
100ng/mL ADA and 100 µg/mL Drug
100ng/mL ADA and 50.0 µg/mL Drug
100ng/mL ADA and 10.0 µg/mL Drug
100ng/mL ADA and 0.0 µg/mL Drug
Acid Dissociation only
2M MgCl2 with acid dissociation
No Dissociation
2M MgCl2 dissociation only

Figure 4: Drug tolerance curves for the conditions indicated in Table 4. Signal to noise values of 1.1 or greater are considered drug tolerant.

Polyclonal Drug Tolerance Summary			
Evaluation	**Specification**	**No Dissociation**	**$MgCl_2$ and Acid Dissociation**
Drug tolerance Comparison of No Dissociation and stepwise $MgCl_2$ and Acid dissociation	Highest tested drug:PC ratio which failed to inhibit 50.0 ng/mL anti-drug Ab below cut point.	<10.0 µg/mL	100 µg/mL
	Highest tested drug:PC ratio which failed to inhibit 100 ng/mL anti-Drug Ab below cut point.	<10.0 µg/mL	>100 µg/mL
	Highest tested drug:PC ratio which failed to inhibit 200 ng/mL anti-Drug Ab below cut point.	50.0 µg/mL	>100 µg/mL

Table 5: Polyclonal Drug Tolerance Summary

CONCLUSIONS

This novel two-step acid and ionic dissociation method uses salt precipitation to aid in removing the newly dissociated drug from the system. The temporary shift to the tertiary and quaternary structures improves the drug tolerance, allowing the remaining free ADAs to be added to a variety of assay types for antibody detection. This method has been successful in achieving drug tolerance without compromising assay sensitivity for the monoclonal assay and increased sensitivity for the polyclonal assay as non-specific binding was decreased.

References

1.Gregor Jordan 1, Alexander Pohler 1, Florence Guilhot 2 et al. - High ionic strength dissociation assay (HISDA) for high drug tolerant immunogenicity testing - Bioanalysis (2020) 12(12), 857866".

www.kcasbio.com