Flow-cytometry based multiparametric profiling of neutrophil function

SCIENCE ACCELERATED

Dmitri Negorev, NatoTeteloshvili, Stefan Hailey
KCAS Bio - PHL, Lower Gwynedd, PA

PURPOSE

Neutrophils consist of phenotypically heterogeneous populations of cells which mediate removal of bacterial and fungal pathogens by phagocytosis and NADP oxidase (nicotinamide adenine dinucleotide phosphate) dependent respiratory burst responses. Recent advances in fluorogenic reagents have improved quantitative measurement of both responses as well as neutrophil subset phenotyping. Inherited neutrophil disorders or chemotherapeutic treatment can compromise neutrophil differentiation and function. Thus, assessment of neutrophil function in patients with immunodeficiencies and undergoing treatment is of increasing importance. While current approaches focus on single parameter measurement of neutrophil function, there is a growing demand for a combined multi-parametric approach, minimizing manipulation of neutrophils and allowing for simultaneous analysis of neutrophil function and phenotyping.

BACKGROUND

Neutrophils are the most abundant innate immune cell with critical anti-microbial functions. Neutrophils follow the leukocyte adhesion cascade to move from the bone marrow to sites of infection or inflammation. Novel aspects occur in the regulation of the leukocyte adhesion cascade during which time they display different phenotypes which results in various neutrophils with distinct properties and specialized functions. Neutrophil surface markers change to facilitate altered functions as the neutrophil matures (Figure 1).

Neutrophils are phagocytes, capable of ingesting microorganisms or particles. For targets to be recognized, they must be coated in opsonins – a process known as antibody opsonization (1). They can internalize and kill many microbes, each phagocytic event resulting in the formation of a phagosome into which reactive oxygen species and hydrolytic enzymes are secreted. The consumption of oxygen during the generation of reactive oxygen species has been termed the “oxidative burst“ (Oxiburst).

Neutrophil dysfunction has been associated with adverse prognosis in a variety of diseases including sepsis, rheumatoid arthritis (RA), systemic lupus erythematosus (SLE), human immunodeficiency virus (HIV), mycobacterium tuberculosis infection and antineutrophil cytoplasmic antibody (ANCA)-associated vasculitis and cancer (2).

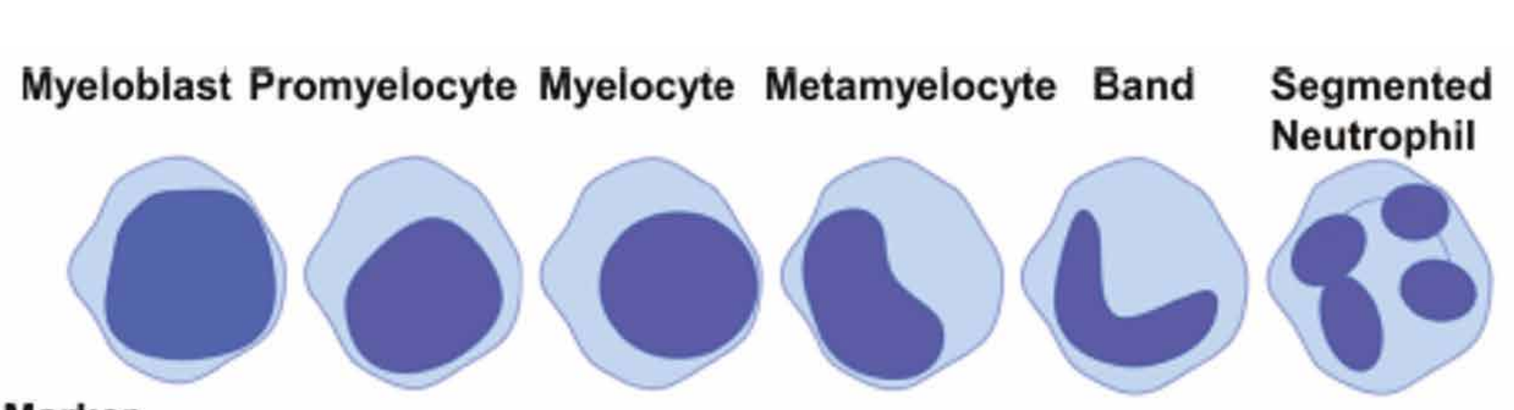
Myeloblast
Promyelocyte
Myelocyte
Metamyelocyte
Band
Segmented Neutrophil

Surface Marker	Myeloblast	Promyelocyte	Myelocyte	Metamyelocyte	Band	Segmented Neutrophil
CD66b	-	+++	+++	++	++	++
CD15	+	+++	+++	+++	+++	++
CD33	+++	+++	++	+	+	+
CD62L	++	++	++	++	++	++
CXCR2	+	+	+	+	+ +	++
CXCR4	++	++	++	++	+	+
CD18	++	+	+++	++	++	++
CD24	-	-	++	++	++	++
CD11b	-	-	+/++	++	++	++
CD11c	-	-	++	++	++	++
CD177	-	-	+	+	+	+
CD16	-	-	-	+	++	+++
CD87	-	-	-	-	++	++
CD10	-	-	-	-	-	++
CD35	-	-	-	-	++	++
HLA-DR	+	-	-	-	-	-
CD34	+	-	-	-	-	-
CD49d	++	++	++	+	-	-

Figure 1. Expression of surface markers during granulopoiesis. Figure illustrates the surface marker expression at each stage of granulopoiesis; myeloblast, promyelocyte, myelocyte, metamyelocyte, band cell and segmented neutrophil. The intensity of the surface marker is shown whereby; low intensity (+), medium intensity (++) and high intensity (+++). Figure is taken from (2).

METHOD AND RESULTS

Peripheral blood samples were collected in sodium heparin tubes from healthy donors and processed immediately. pH sensitive indicating fluorescent Escherichia coli (E. coli) particles were opsonized and incubated with whole blood samples for detection of phagocytic cells (Figure 2). In a separate assay whole blood samples were first labeled with Dihydrorhodamine-123 and stimulated with opsonized E. coli to measure oxidative burst potential. Prior to acquisition, cells from both functional assays were stained with a fixable live/dead dye and an antibody cocktail including extracellular lineage and maturation markers (Figure 3, A), then red blood cells were lysed, samples were washed and acquired on a BD LSRFortessa. Method optimization studies were performed comparing peripheral blood processed immediately (baseline) with blood stored at room temperature for 24 and 48-hours post sample collection (Figure 4). Combined assays in one test tube (Figure 5).Trucount absolute bead tubes were utilized to enumerate the white blood cell (WBC) counts for each sample.

A

Oxiburst

1. 37°C background control
2. 37°C experimental (Opsonized *E.coli*)
3. 37°C positive control (PMA)
4. 4°C experimental (Opsonized *E.coli*)

Phagocytosis

1. 37°C Background control
2. 37°C Experimental (Opsonized pHrodo)
3. 4C° Experimental (Opsonized pHrodo)

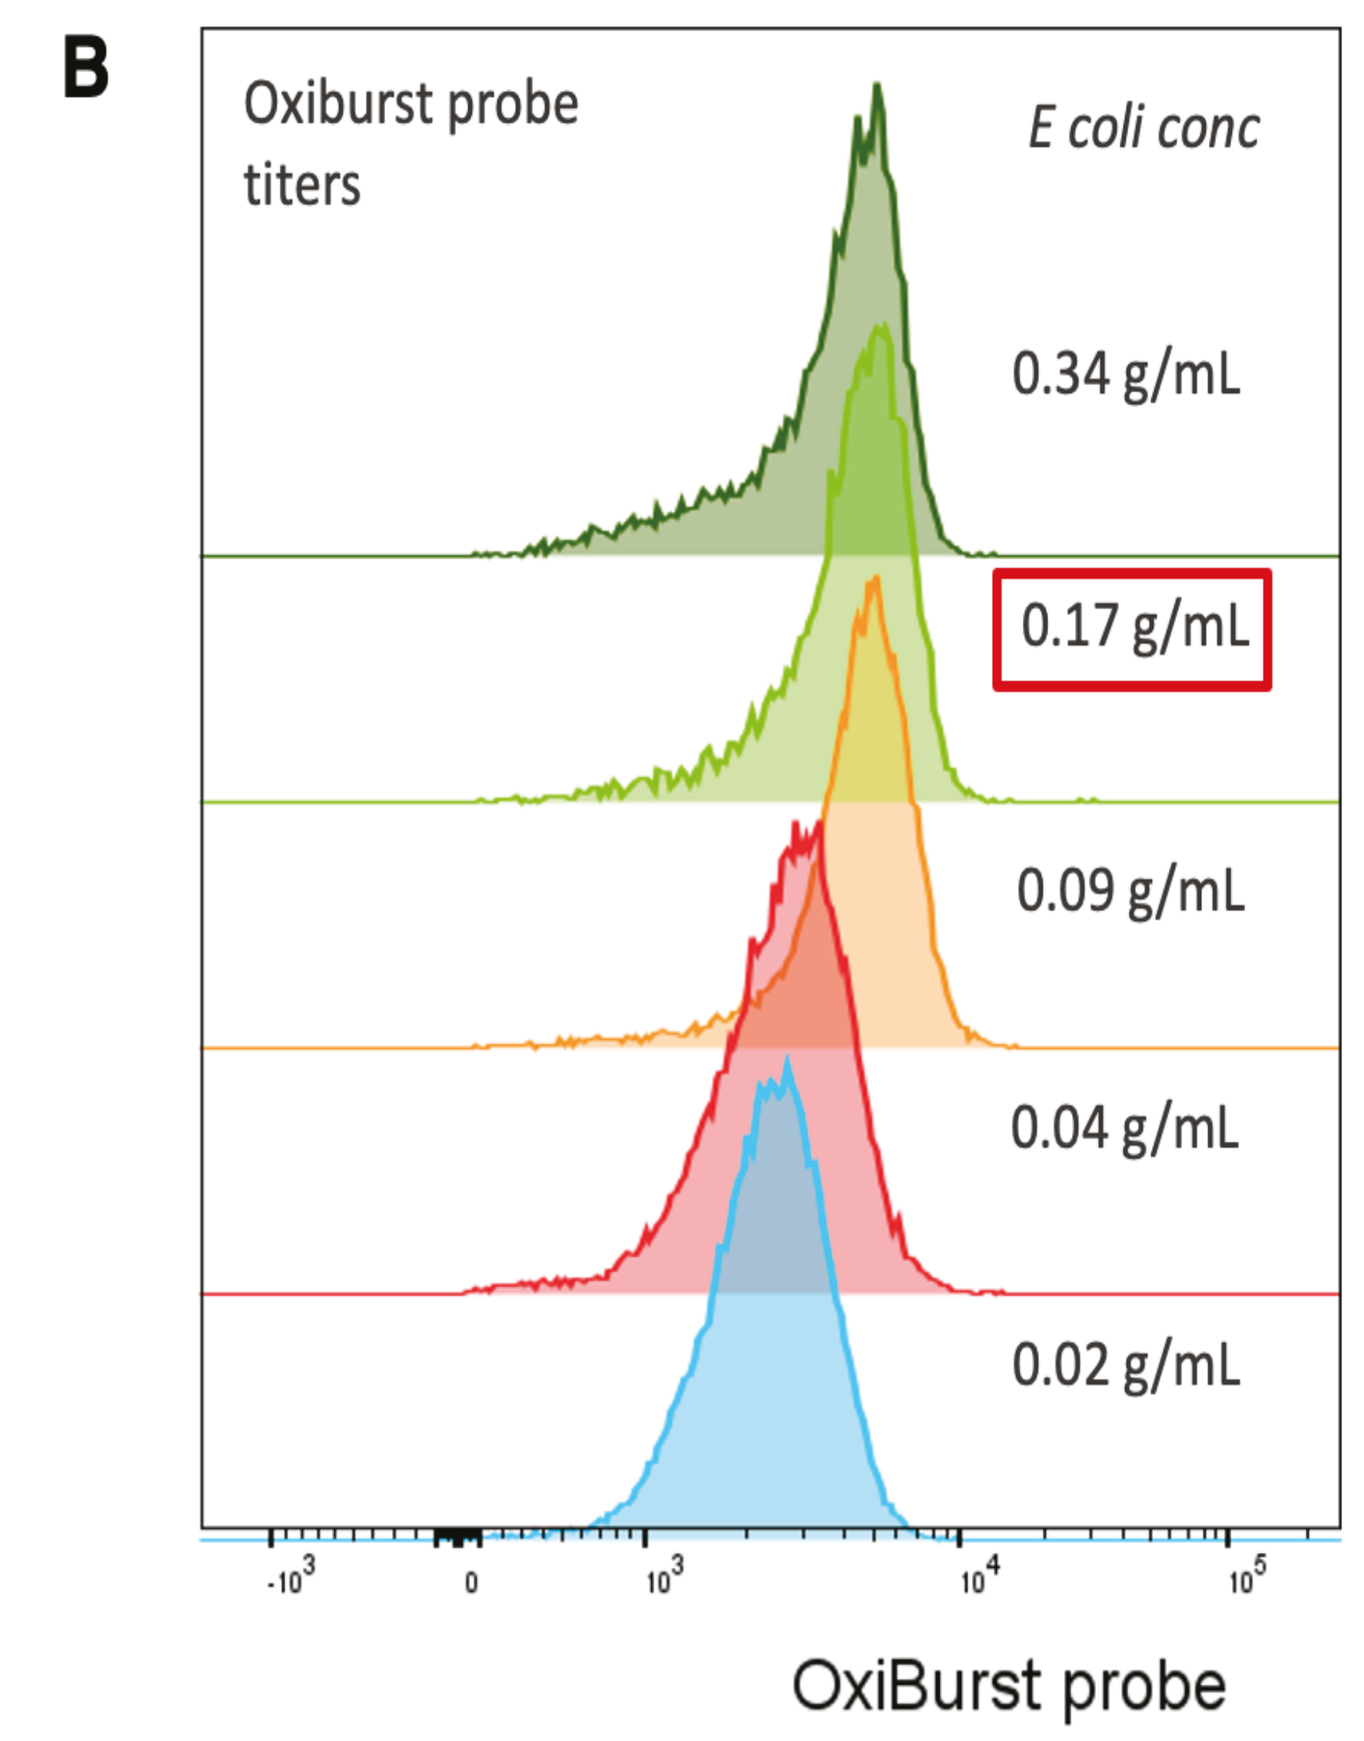
B
Oxiburst probe titers
E coli conc
0.34 g/mL
0.17 g/mL
0.09 g/mL
0.04 g/mL
0.02 g/mL
OxiBurst probe

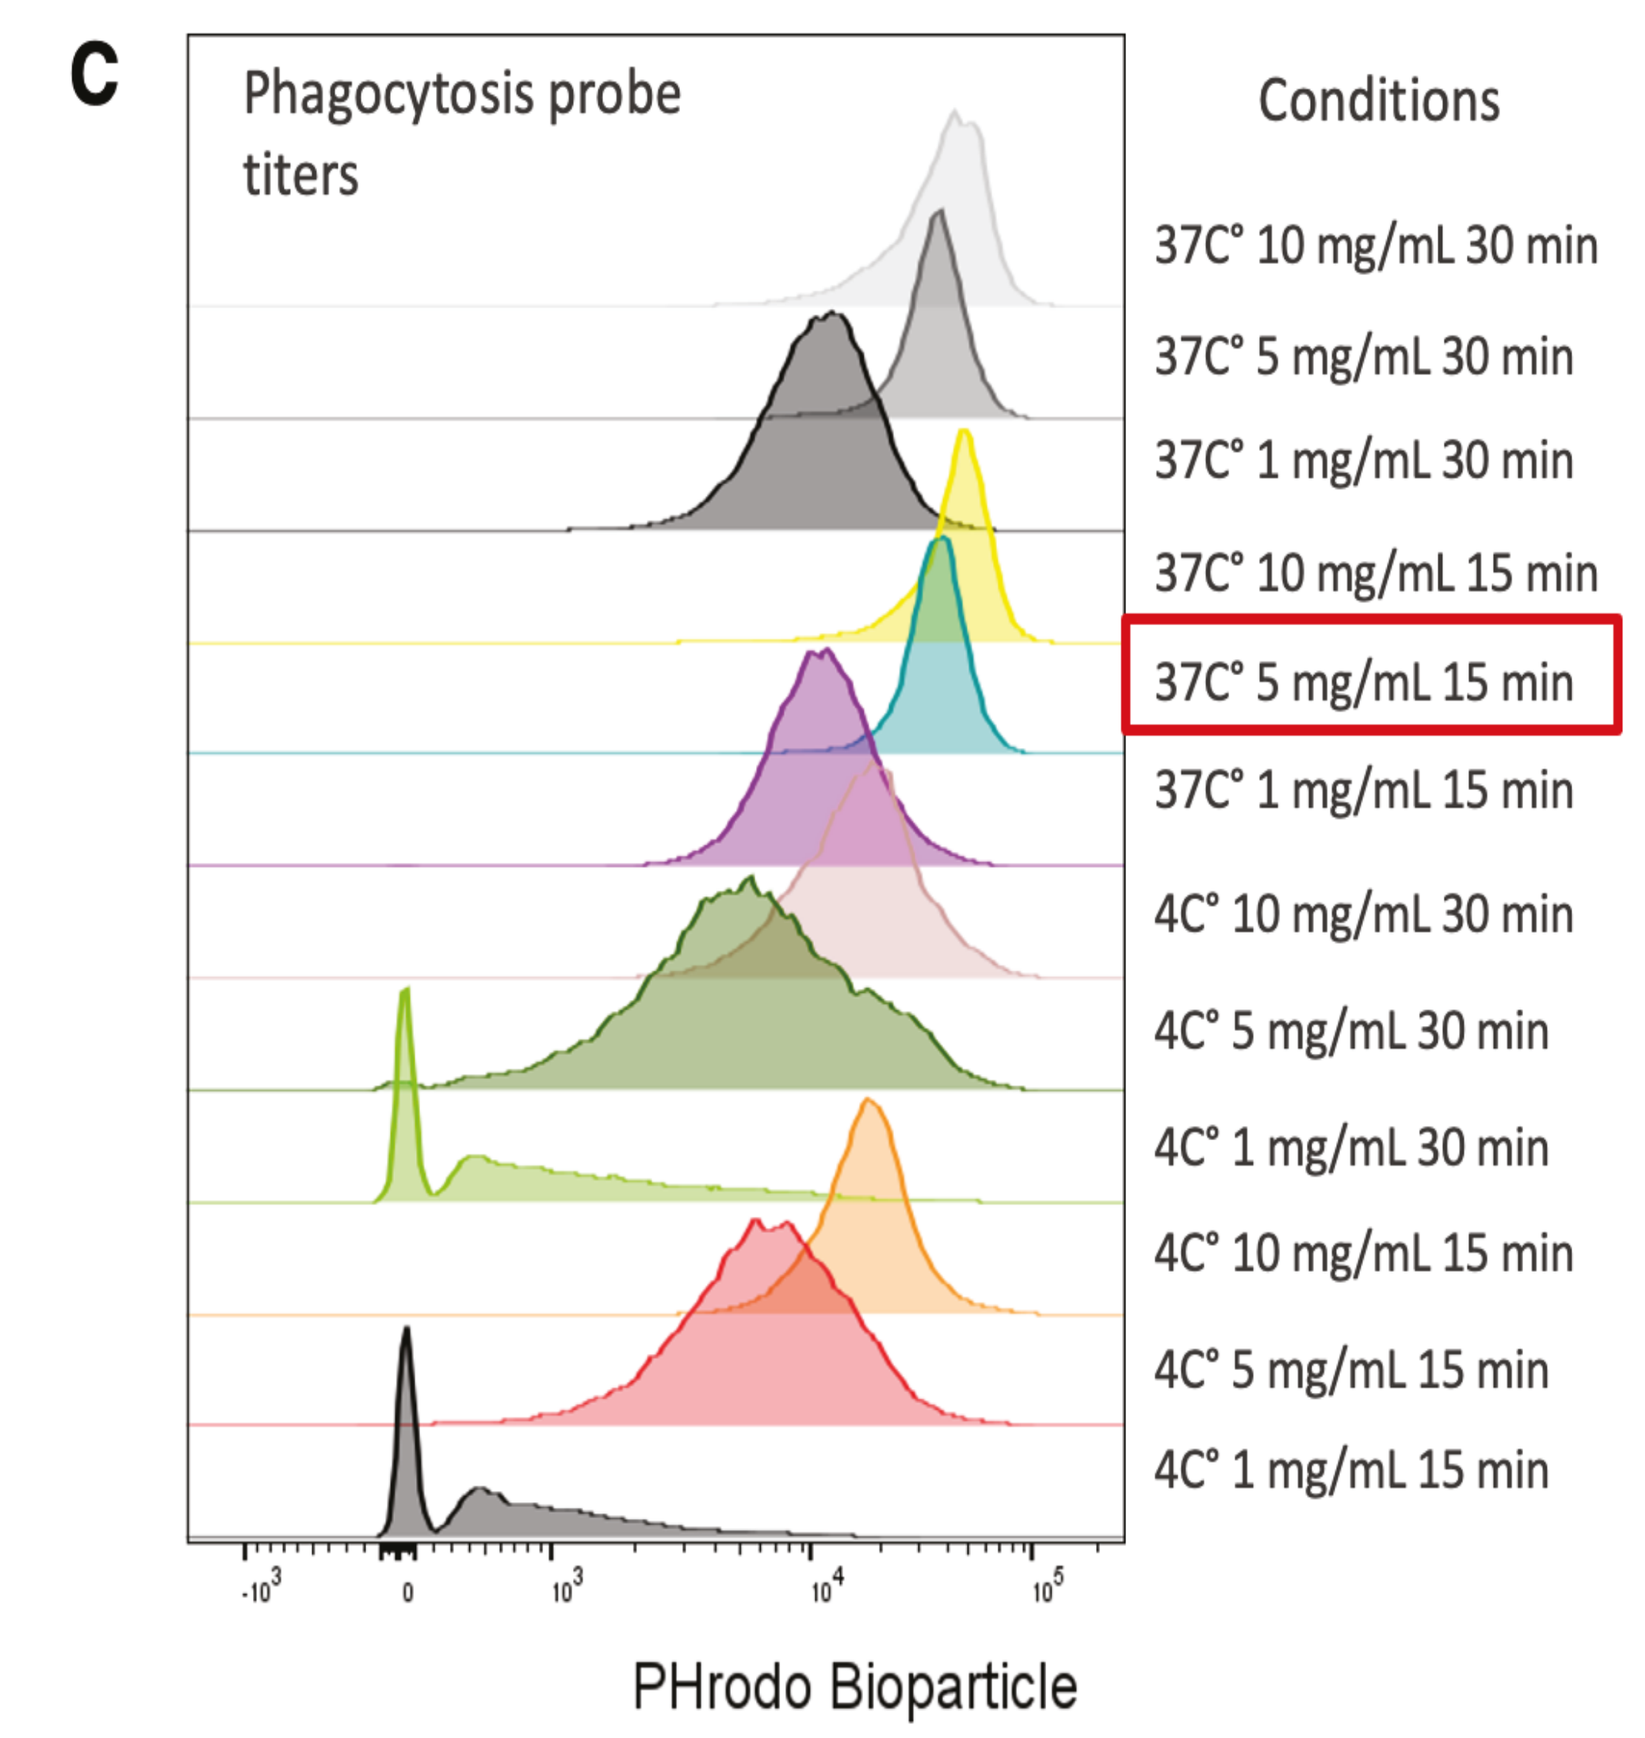
C
Phagocytosis probe titers
Conditions
37C° 10 mg/mL 30 min
37C° 5 mg/mL 30 min
37C° 1 mg/mL 30 min
37C° 10 mg/mL 15 min
37C° 5 mg/mL 15 min
37C° 1 mg/mL 15 min
4C° 10 mg/mL 30 min
4C° 5 mg/mL 30 min
4C° 1 mg/mL 30 min
4C° 10 mg/mL 15 min
4C° 5 mg/mL 15 min
4C° 1 mg/mL 15 min
PHrodo Bioparticle

Figure 2: Experimental conditions (A) and titrations for: Oxiburst (B) and Phagocytosis (C) assays. Red frame represents conditions selected for father analysis.

A

Antibody
CD10
CD19
CD56
CD3
CD14
Siglec8
CD15
CD11b
CD62L
CD184
CD45
CD66b
CD16
PHprobe
Oxiburst
Live/Dead

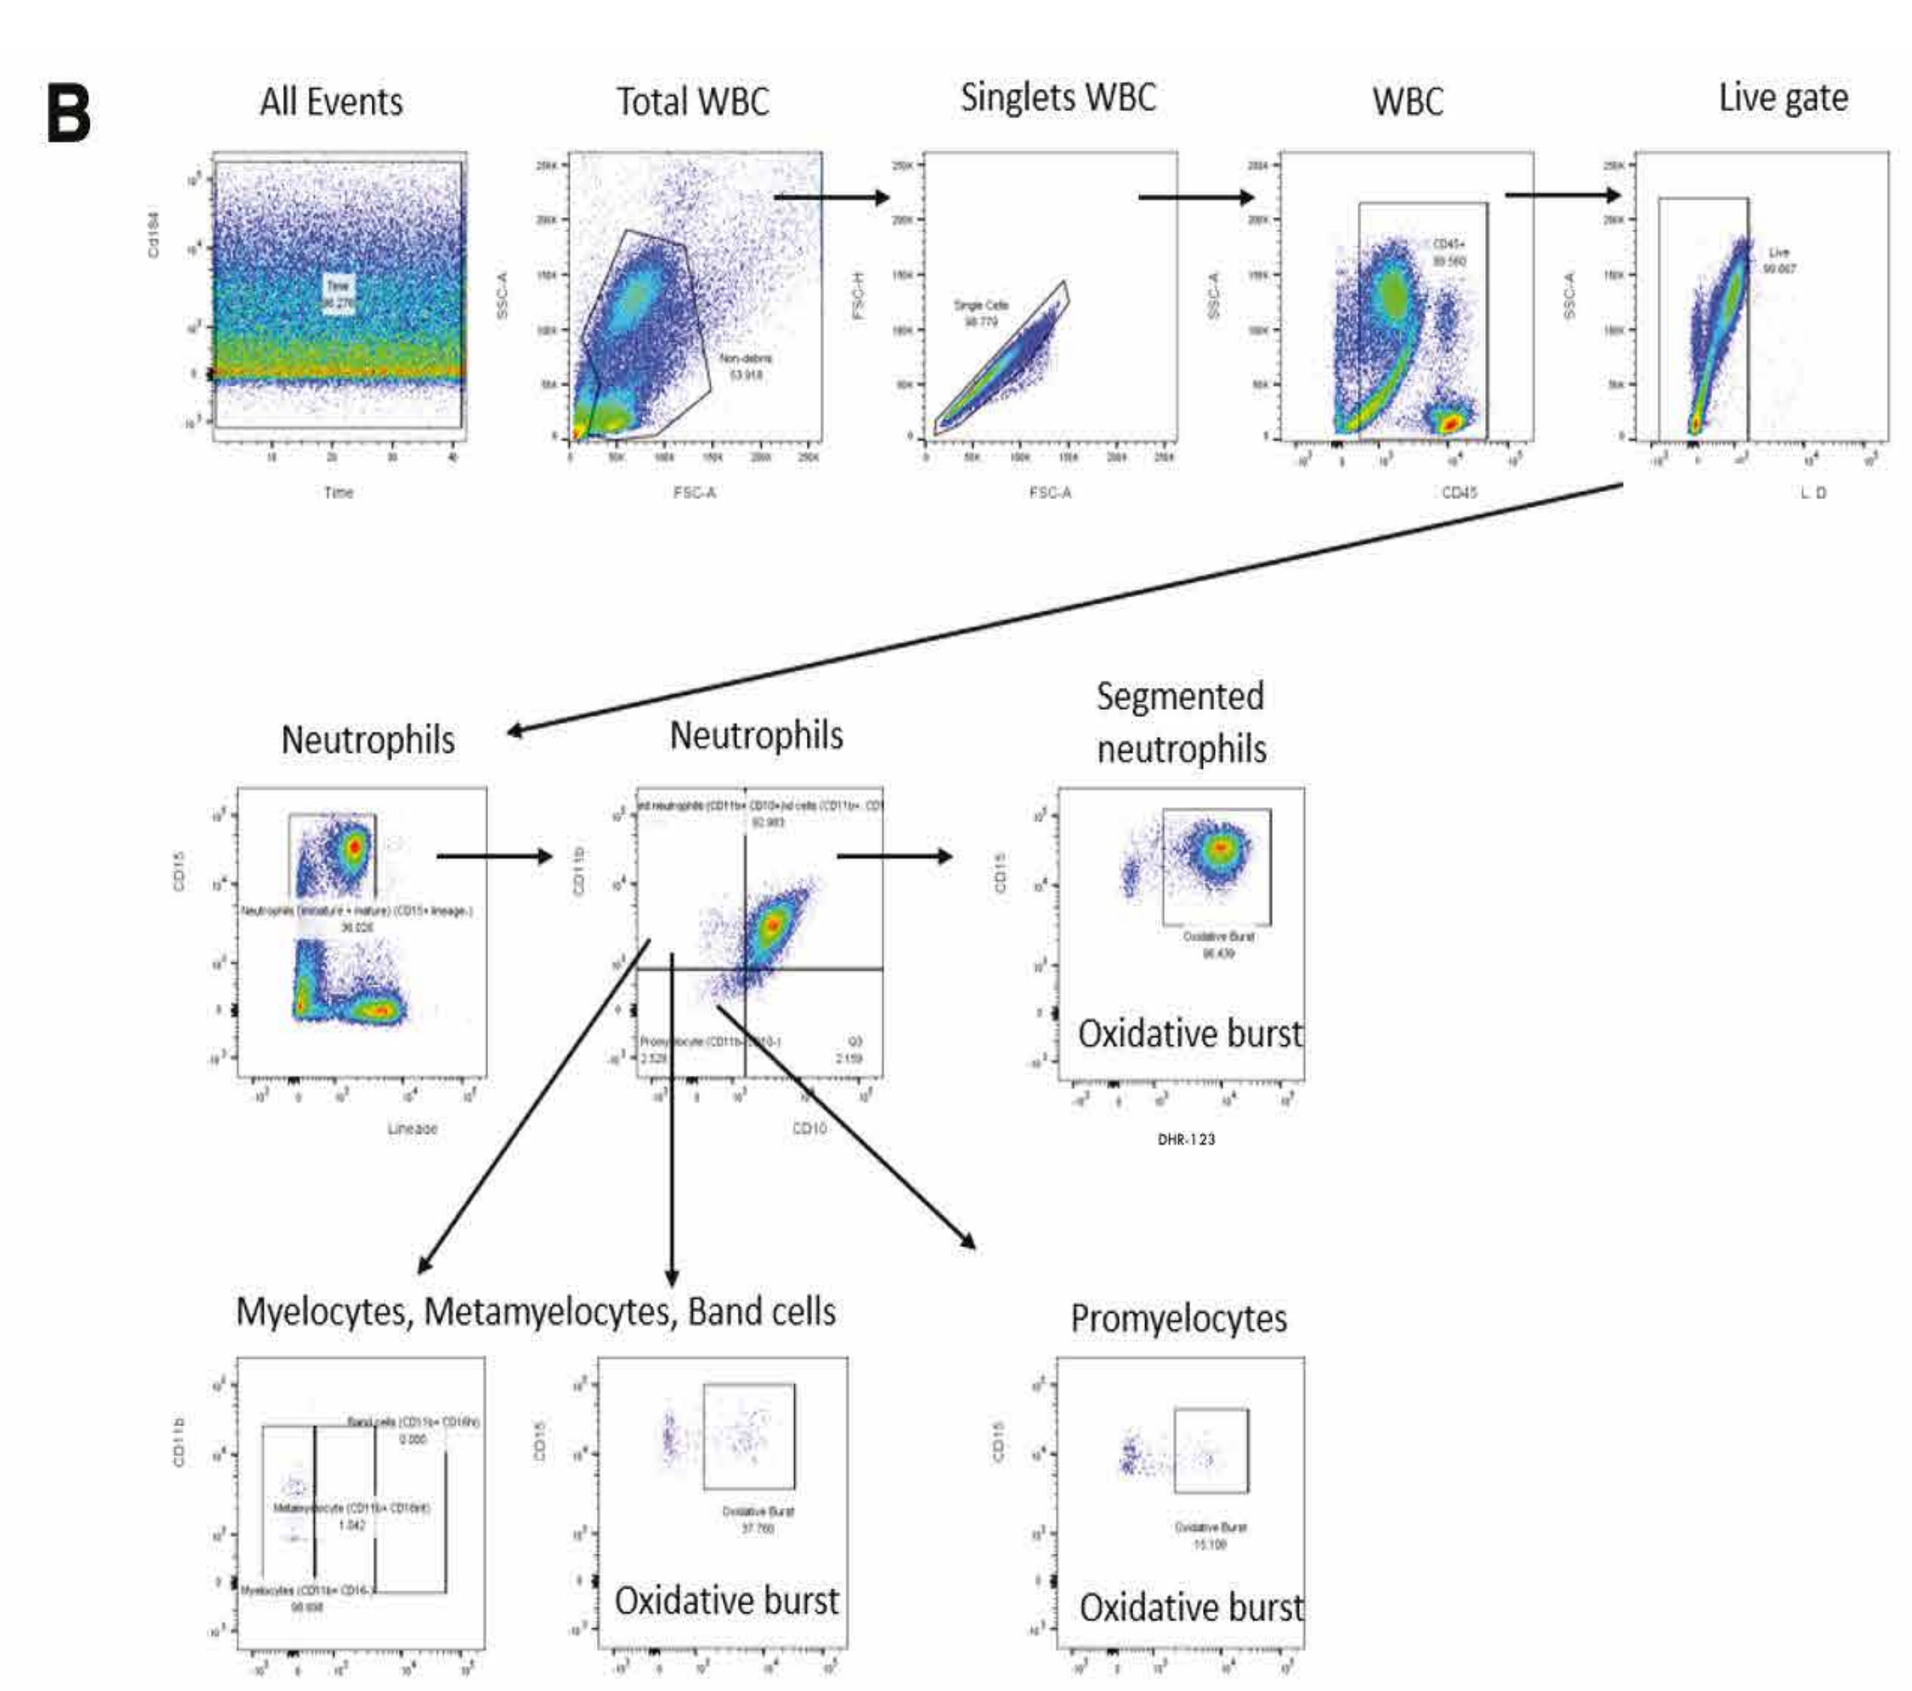
B
All Events
Total WBC
Singlets WBC
WBC
Live gate
Neutrophils
Neutrophils
Segmented neutrophils
Oxidative burst
Myelocytes, Metamyelocytes, Band cells
Oxidative burst
Promyelocytes
Oxidative burst

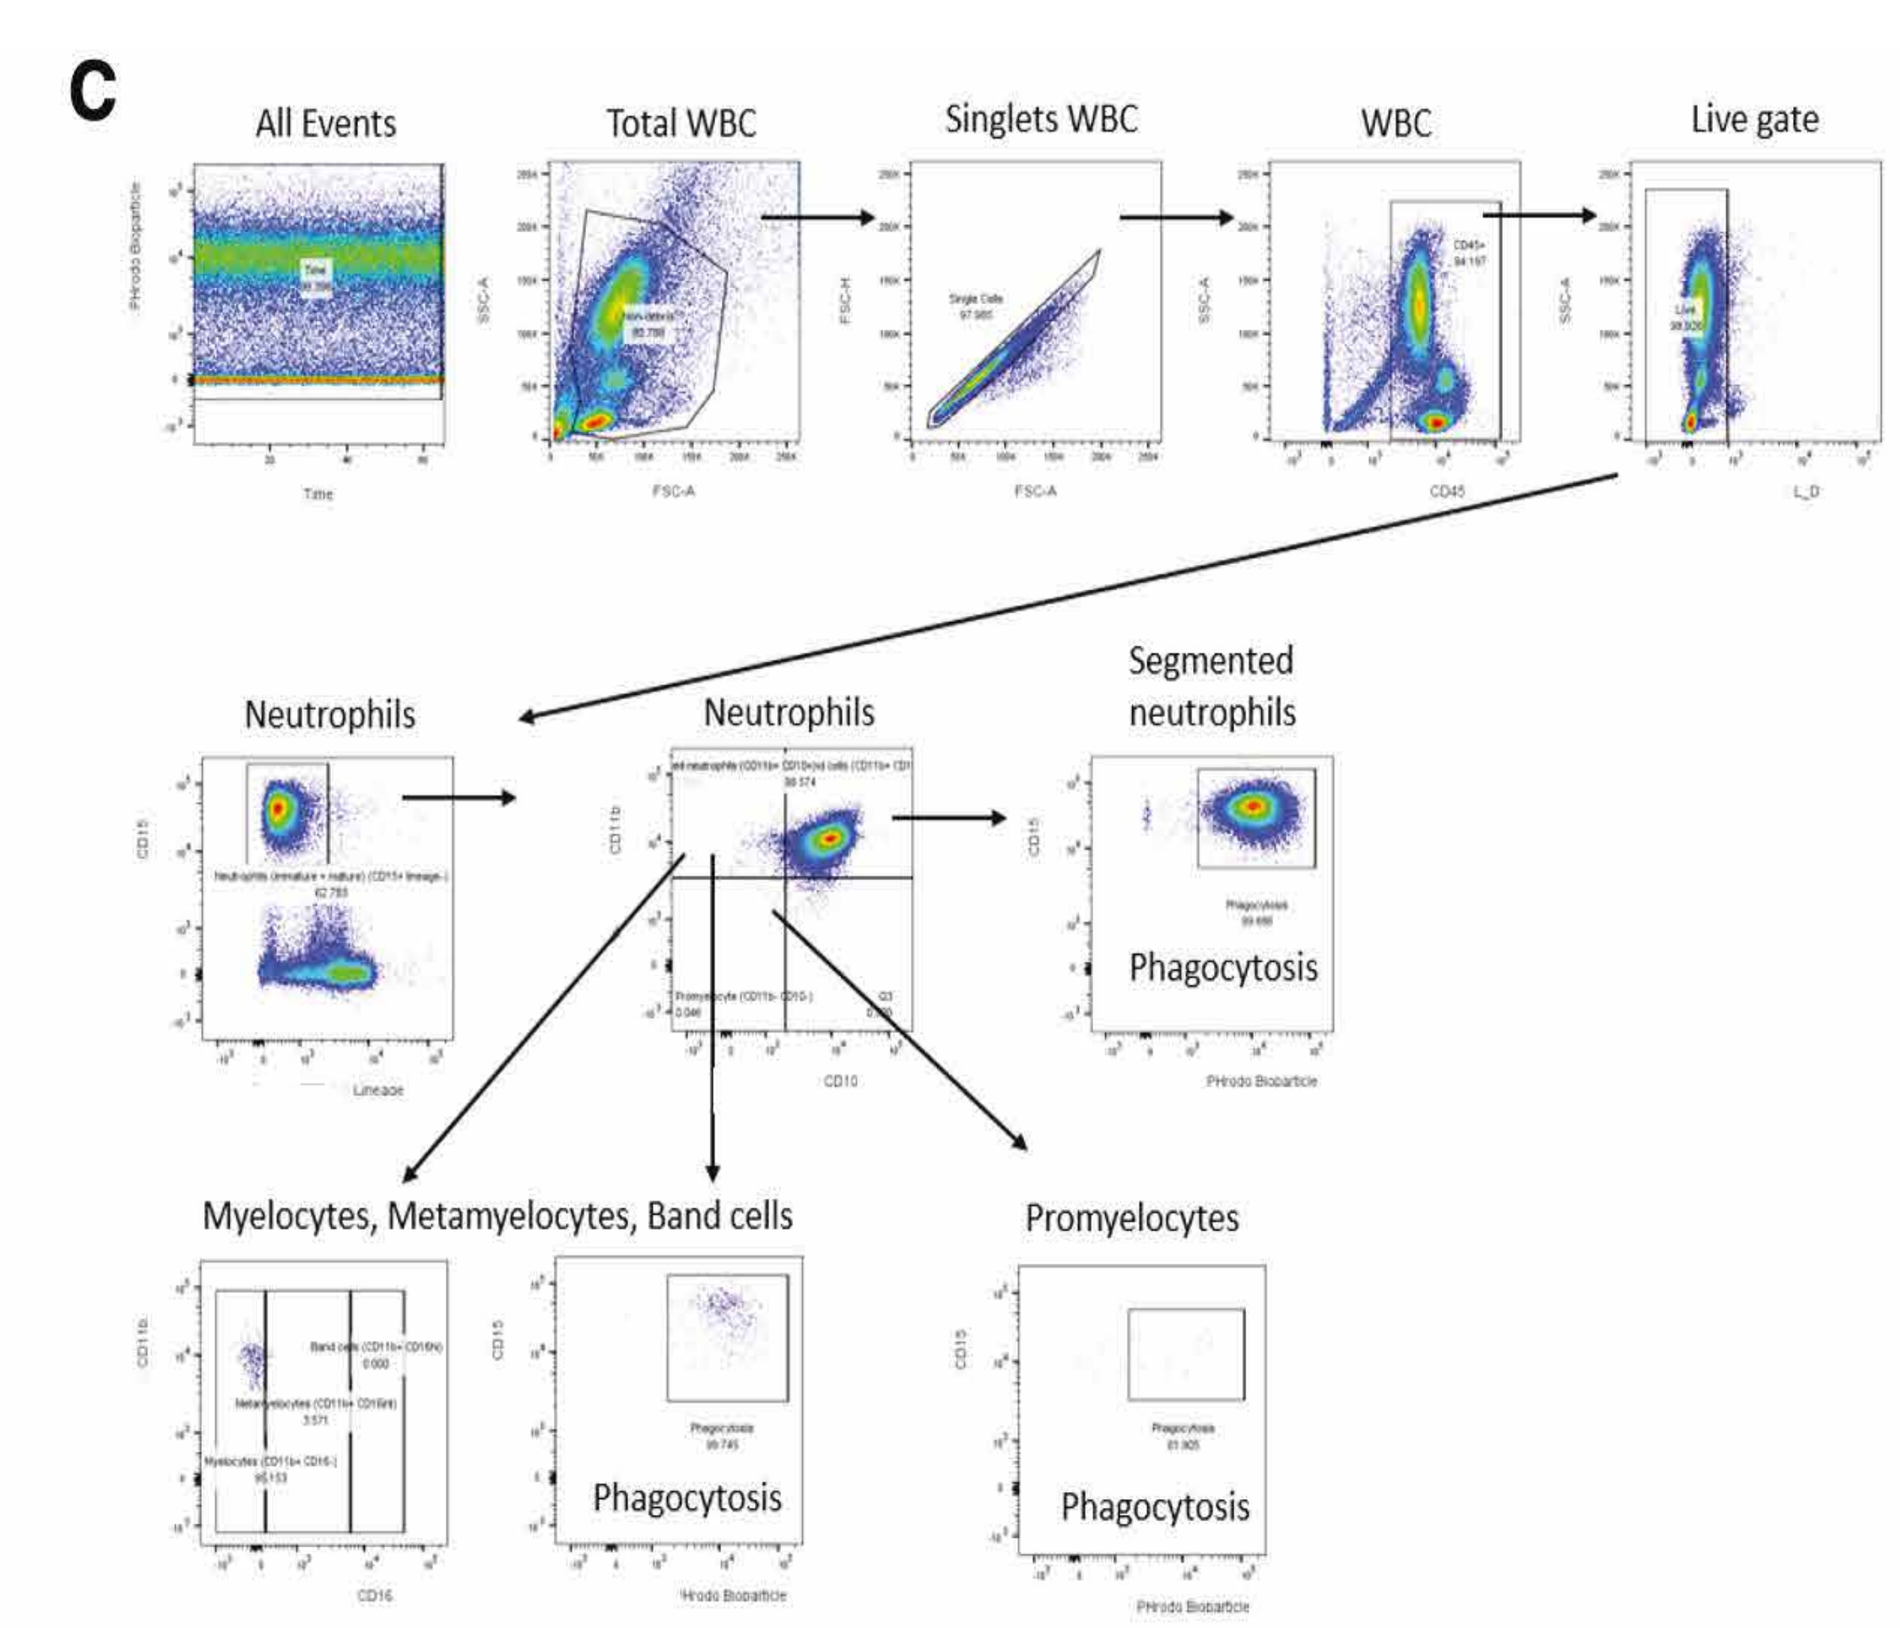
C
All Events
Total WBC
Singlets WBC
WBC
Live gate
Neutrophils
Neutrophils
Segmented neutrophils
Phagocytosis
Myelocytes, Metamyelocytes, Band cells
Phagocytosis
Promyelocytes
Phagocytosis

Figure 3: Immunophenotyping Panel design (A) and Gating strategy for Oxiburst (B) and Phagocytosis (C) assays. All antibodies were first titrated. Neutrophils selected based on positive expression of CD15 and negative gate for lineage markers: CD3, CD19, CD14, CD56, Siglec8. Mature and immature neutrophils characterized based on CD11b and CD10 to study segmented neutrophils and Promyelocytes. Functional assays are analyzed within segmented neutrophils and CD10-CD11b+ neutrophils (promyelocytes, myelocytes, metamyelocytes and band cells).

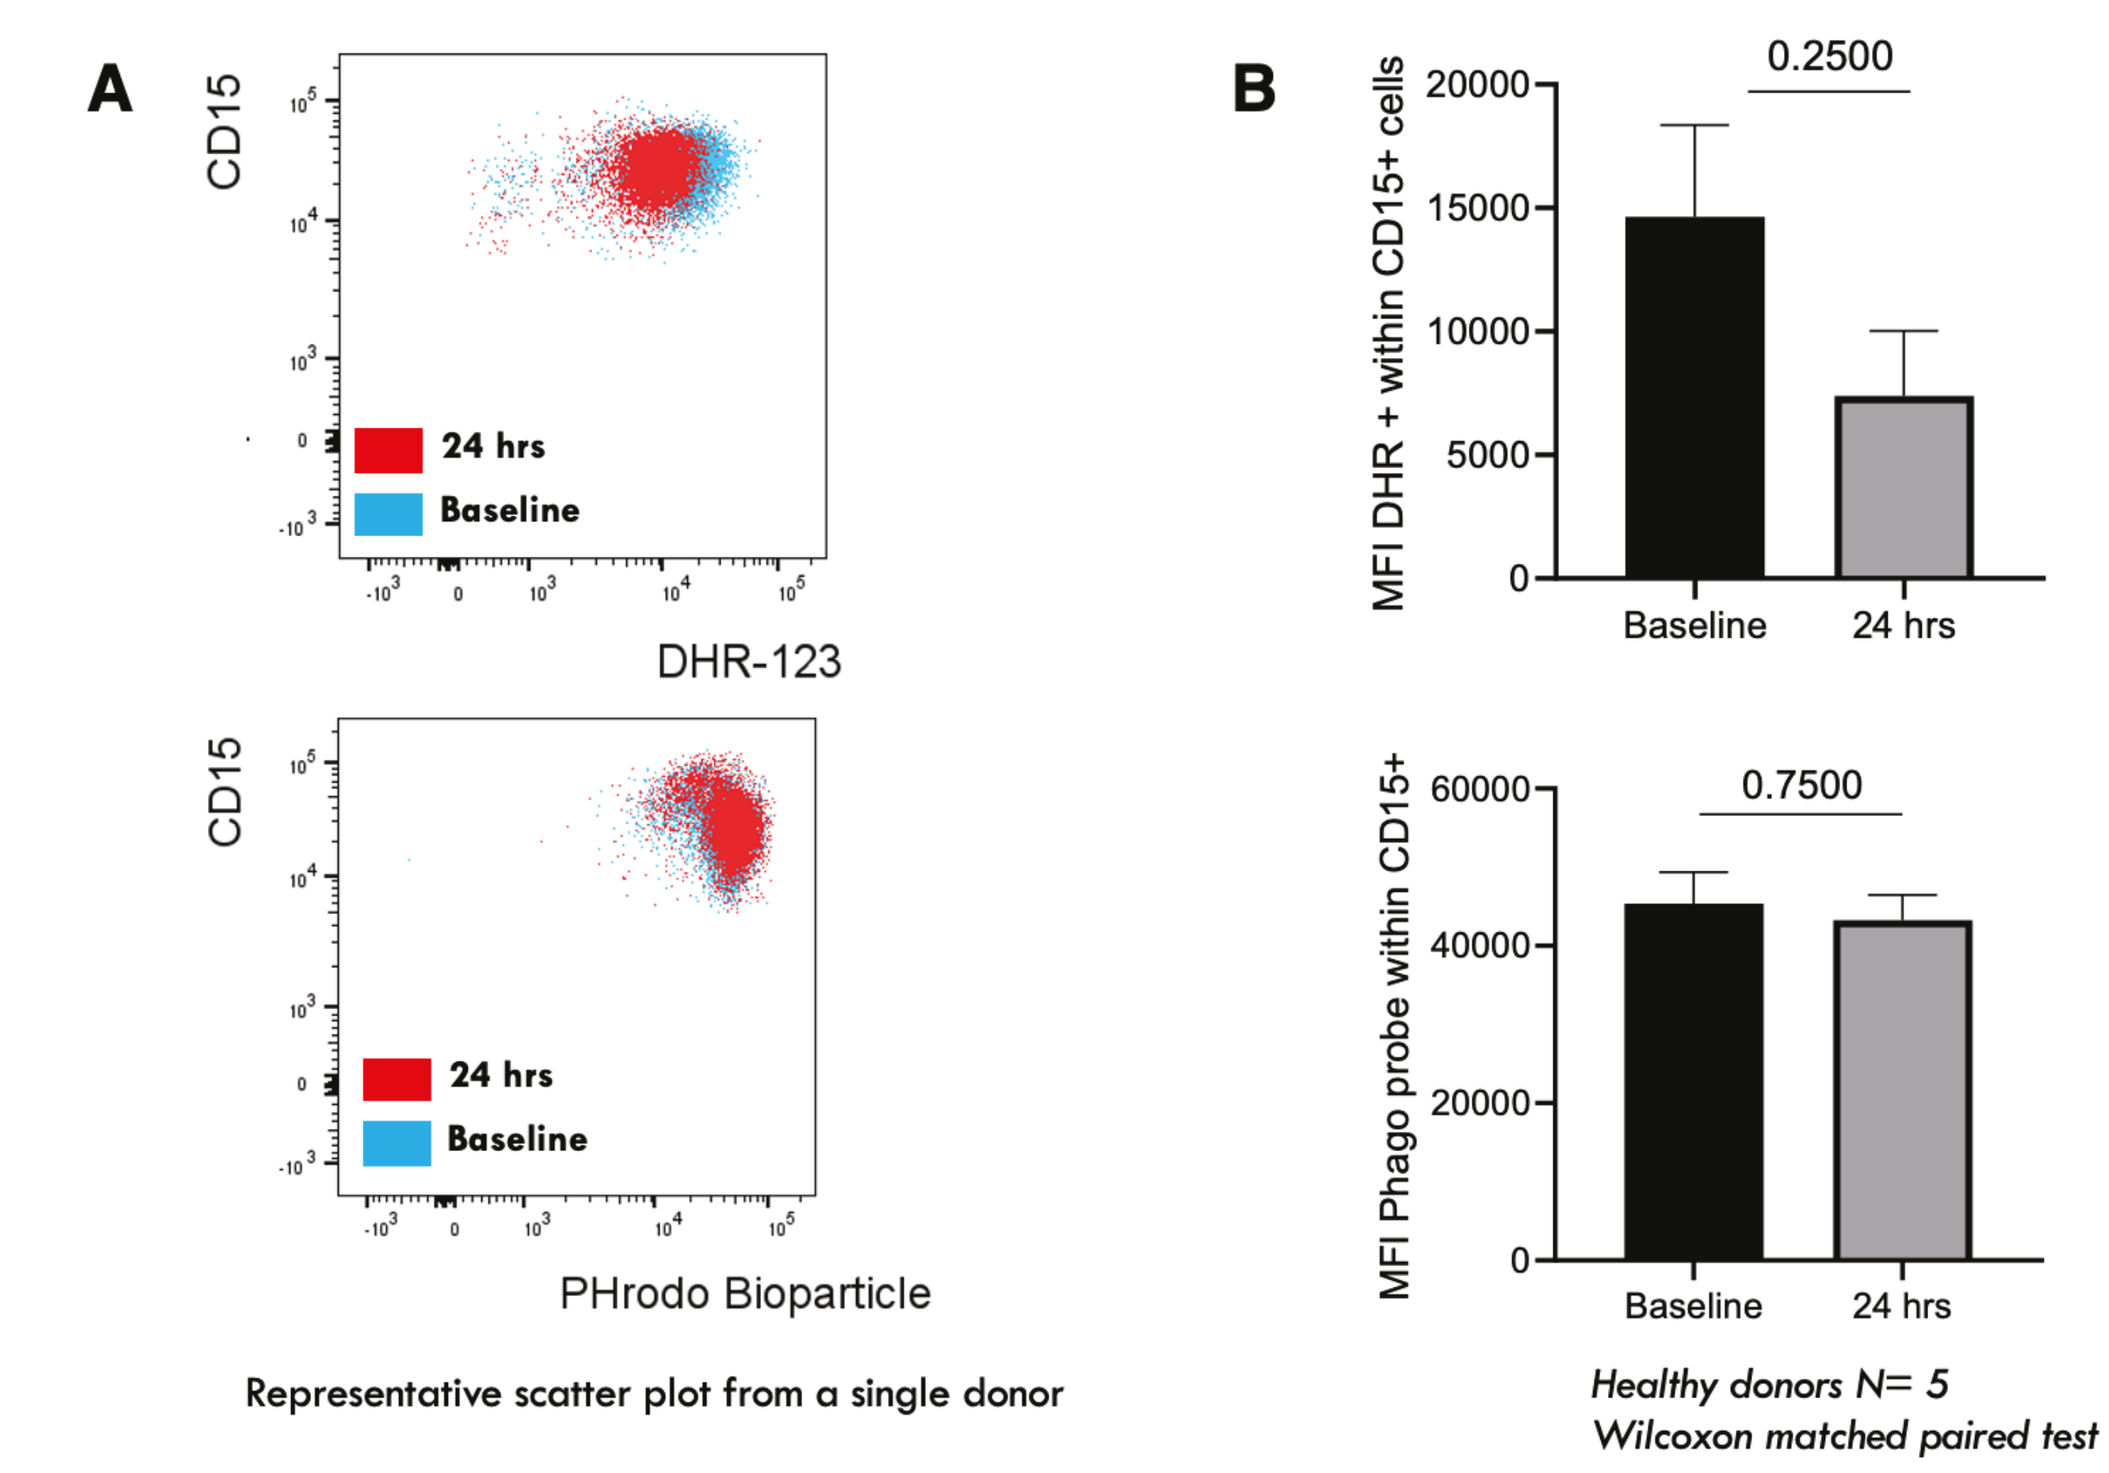
A
CD15
24 hrs
Baseline
DHR-123
CD15
24 hrs
Baseline
PHrodo Bioparticle
Representative scatter plot from a single donor
B
0.2500
MFI DHR + within CD15+ cells
20000
15000
10000
5000
0
Baseline
24 hrs
0.7500
MFI Phago probe within CD15+
60000
40000
20000
0
Baseline
24 hrs
Healthy donors N= 5
Wilcoxon matched paired test

Figure 4: Stability; Whole blood from 5 Healthy Donors was treated and IP analyzed. Only gate for CD15 positive cells is shown (A) and statistical analysis of MFI values (B). (48-hour data not shown as blood was coagulating).

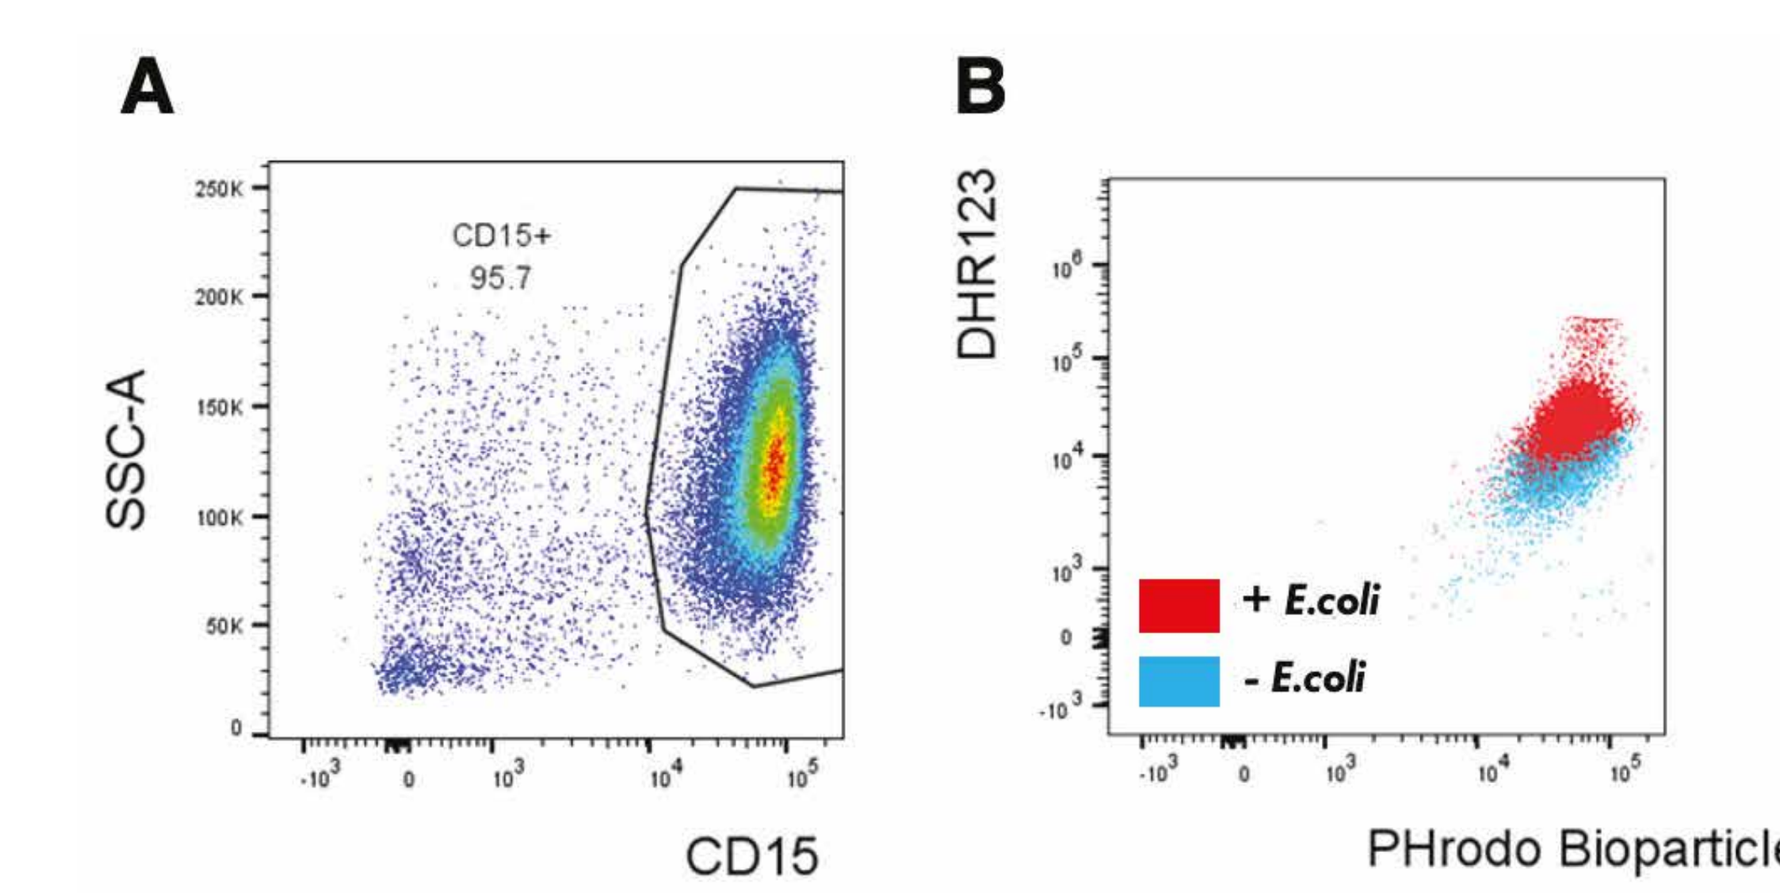
A
CD15+
95.7
SSC-A
CD15
B
DHR123
+ E.coli
- E.coli
PHrodo Bioparticle

Figure 5: Method optimization to combine Oxiburst and Phagocytosis assays. Healthy Donor whole blood was treated and IP analyzed. Only gate for CD15 positive cells is shown (A) and combined detection of DHR123 and PHrodo Bioparticle (B).

CONCLUSIONS

• We have shown reproducible detection and quantification of phagocytosis and oxidative burst activity of neutrophils.

• Indirect detection of the phagocytosis and oxidative burst activity via fluorogenic probes remained interpretable at 24-hour post collection, but not the 48-hour timepoint. An immunophenotyping procedure used to lyse erythrocytes and stain extracellular neutrophil markers also allowed us to discriminate important populations at both baseline and 24-hour timepoints, but not the 48-hour timepoint.

• It is feasible to combine Oxiburst, Phagocytosis and immunophenotyping assays in one test tube.

• Neutrophil phagocytosis and Oxidative burst assays combined with immunophenotyping allow for a robust and rapid assessment of functional status of neutrophils in the whole blood to assess disease state.

REFERENCES

1. Edwards SW (1994). Biochemistry and physiology of the neutrophil. Cambridge University Press. p. 6.
2. McKenna E (2021). Neutrophils: Need for Standardized Nomenclature. Front Immunol.

www.kcasbio.com